Bosentan Prevents Occurrence But Does Not Speed Healing of Digital Ulcers in Patients with Systemic Sclerosis (SSc)

James R. Seibold[1], Christopher P. Denton[2], Daniel E. Furst[3], Marco Matucci-Cerinic[4], Maureen D. Mayes[5], Adele Morganti[6], Fabrice Kramer[6], Joseph H. Korn[7] for the RAPIDS-2 Investigators.

[1]University of Michigan Scleroderma Program, Ann Arbor, MI, USA; [2]Royal Free Hospital, London, United Kingdom; [3]UCLA, Los Angeles, CA, USA; [4]University of Florence, Florence, Italy; [5]University of Texas-Houston, Houston, TX, USA; [6]Actelion Pharmaceuticals Ltd, Allschwil, Switzerland; [7]Boston University, Boston, MA, USA

ABSTRACT

Objective

Digital tip ulcers (DTU) are an important source of disease-related morbidity in SSc for which there are no effective oral therapies. Bosentan, a dual endothelin receptor antagonist, was associated with a significant reduction in new DTU in 122 patients with SSc treated over 16 weeks (Korn JH. *Arthritis Rheum* 2004; 50: 3985-3993). We report a randomized clinical trial (RCT) with bosentan in which we sought to confirm effects on prevention and to measure potential effects on healing of DTU in SSc.

Methods

188 subjects were studied in 41 centers in North America and Europe. Eligibility included a diagnosis of SSc and at least one recent active DTU. In subjects with more than one active DTU, a "cardinal ulcer" was identified by subject and investigator based on location, clinical impact and amenability to healing. Subjects received bosentan at 62.5 mg bid for 4 weeks then 125 mg bid for 20 to 32 weeks or placebo (PBO) in 1:1 blinded randomization. All DTUs were assessed at 4 weekly intervals. Co-primary endpoints were time to complete healing of the cardinal ulcer and the number of new DTUs that developed during 24 weeks of study. Secondary endpoints included SHAQ-DI and safety.

Results

Subjects were well balanced between bosentan and PBO by age, gender, SSc classification, smoking history and concomitant medications. Total number of new ulcers over 24 weeks was 1.9 ± 0.2 on bosentan vs. 2.7 ± 0.3 on PBO ($p = 0.035$, Pitman permutation). Measures of healing were comparable between bosentan and PBO including time to healing of cardinal ulcer, time to healing of all DTUs and percent with complete healing (36.8% bosentan vs 39.3% PBO, $p = 0.76$). Improvement in SHAQ-DI trended in favor of bosentan (-0.11) vs PBO (-0.05) at 12 weeks but was not significant ($p = 0.312$). SHAQ "dressing" improvement was significant for bosentan over PBO at 24 weeks ($p = 0.033$) and trended in favor of drug for "eating" at 24 weeks ($p = 0.098$). SHAQ VAS scales were improved on bosentan for pain at 12 weeks ($p = 0.034$). SAE's were uncommon. ALT/AST > 3X ULN was more frequent on bosentan (10.5%) than PBO (1.1%).

Conclusions

This second large RCT confirms that bosentan reduces the occurrence of new DTUs in patients with SSc and that this effect is associated with reduced pain and improved hand function. Bosentan therapy does not appear to be effective for Raynaud's phenomenon or in facilitating healing of active DTUs. These data suggest that bosentan has an important clinical effect on peripheral vascular integrity and function.

Background and Rationale

- Progressive occlusive fibrotic vasculopathy is the hallmark of systemic sclerosis (scleroderma; SSc)
- Vascular syndromes in scleroderma include pulmonary arterial hypertension (PAH), renal involvement, Raynaud's phenomenon and ischemic digital ulcers
- Digital ulcers occur in around 50% of patients and are responsible for pain, limitation of hand function and tissue loss
- There are no effective therapies for digital ulcerations
- Endothelin-1 and its receptors, notably the ET_B receptor, are overexpressed in scleroderma
- The vasoconstrictive and pro-proliferative effects of ET-1 contribute to the vasculopathy of scleroderma
- Bosentan (Tracleer®) is a dual ET-1 receptor antagonist of proven effectiveness in PAH WHO Class III and IV including PAH secondary to SSc
- The role of bosentan in the prevention of new DUs in SSc was presented in an earlier RCT, RAPIDS-1 (Korn JH. *Arthritis Rheum* 2004; 50: 3985-3993)
- The present study (RAPIDS-2) was designed to 1) substantiate the potential effect of bosentan on prevention of new digital ulcers in SSc and 2) measure the potential benefit of bosentan on healing of existing ulcers

METHODS

- A randomised, placebo-controlled, double-blind, prospective, multi-centre study.

RAPIDS-2 – Study design

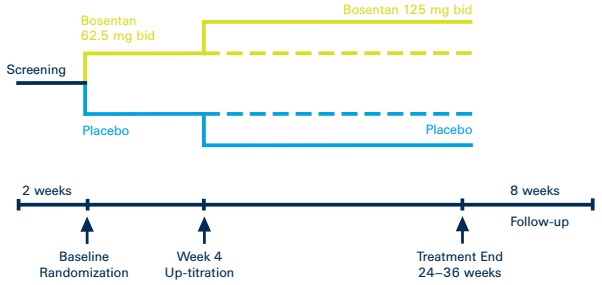
Bosentan 62.5 mg bid
Bosentan 125 mg bid
Screening
Placebo
Placebo
2 weeks
8 weeks
Follow-up
Baseline Randomization
Week 4 Up-titration
Treatment End 24–36 weeks

- Diagnosis of SSc by ACR classification criteria
- At least one active digital ulcer at baseline
- In subjects with more than one active digital ulcer, a "cardinal" ulcer was identified based on severity, location and amenability to healing.
- Co-primary endpoints were time to complete healing of cardinal ulcer and number of new digital ulcers over 24 weeks.
- Secondary endpoints included SSc Health Assessment Questionnaire (SHAQ), safety and tolerability.

RESULTS

Patient demographics and disease characteristics

	Placebo	Bosentan
No. patients	90	98
Males (%)	20	22.4
Age (yrs)	50.7	48.4
Weight (kg)	66.5	64.7
Caucasian (%)	83	87
Black (%)	6	7
Diffuse SSc (%)	42.2	39.8
Time from Diagnosis of SSc (yrs)	8.7	8.7
No. DU at baseline	3.6	3.7

Results: Healing

Time to complete healing of the cardinal ulcer up to week 24

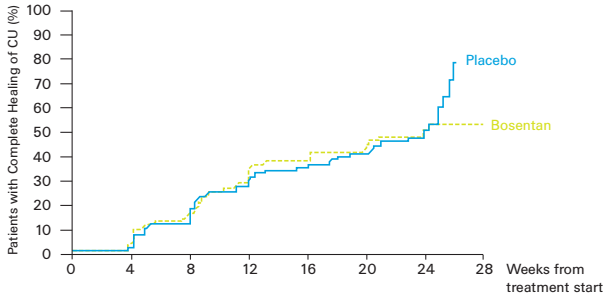
Patients with Complete Healing of CU (%)
100
90
80
70
60
50
40
30
20
10
0
0
4
8
12
16
20
24
28
Weeks from treatment start
Placebo
Bosentan

Patients at risk								
Bosentan	98	95	84	68	62	57	31	4
Placebo	90	89	76	64	59	54	34	0

Results: Prevention

New DUs at week 12 and week 24

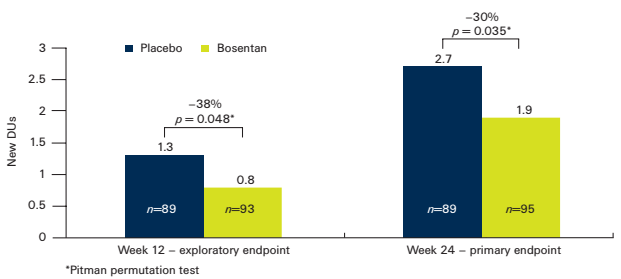
Placebo
Bosentan
New DUs
3
2.5
2
1.5
1
0.5
0
–38%
p = 0.048*
1.3
0.8
n=89
n=93
–30%
p = 0.035*
2.7
1.9
n=89
n=95
Week 12 – exploratory endpoint
Week 24 – primary endpoint

*Pitman permutation test

New DUs up to week 24 by pre-defined co-variables (placebo-corrected)

	Bosentan	Placebo
All patients	95	89
SSc type		
Diffuse	37	37
Limited	58	52
Time from DU to randomisation		
≤ 5 years	54	49
> 5 years	41	39
Systemic antibiotics for DU		
Yes	16	11
No	76	77
Local treatments for DU		
Yes	33	24
No	61	64
Change in treatment of Raynaud's phenomenon		
Yes	22	19
No	72	69
Smoking at baseline		
Never smoked	58	49
Previous smoker	22	20
Current smoker	12	19

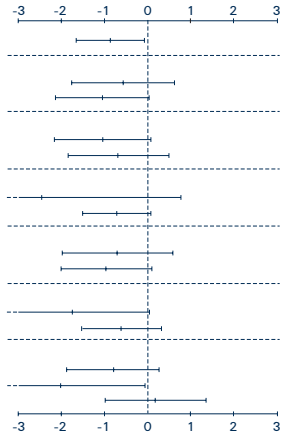
-3
-2
-1
0
1
2
3

New DUs up to week 24 by number of baseline DUs

		Placebo	Bosentan	Δ
Bas DU≤3	All treated *(n)*	60	57	
	No. new DUs	1.9	1.4	-26%
Bas DU>3	All treated *(n)*	29	36	
	No. new DUs	4.4	2.3	-48%

SHAQ items change from baseline to week 12 and week 24

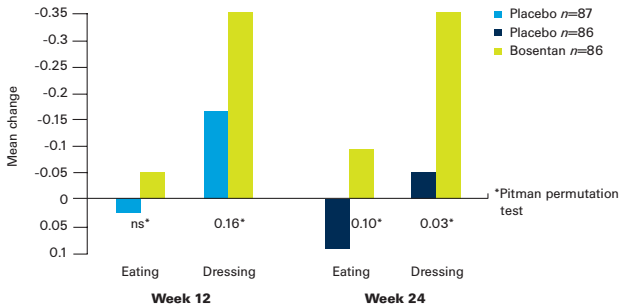
Mean change
-0.35
-0.3
-0.25
-0.2
-0.15
-0.1
-0.05
0
0.05
0.1
Placebo n=87
Placebo n=86
Bosentan n=86
ns*
0.16*
0.10*
0.03*
Eating
Dressing
Eating
Dressing
Week 12
Week 24
*Pitman permutation test

Pain

SHAQ VAS scales were improved on bosentan at 12 weeks ($p = 0.034$)

Safety

Overall bosentan was well tolerated and the safety profile was consistent with earlier RCTs of bosentan. All transaminase elevations were reversed on treatment discontinuation of bosentan

CONCLUSIONS

- Bosentan appears effective in preventing the occurrence of digital tip ulcerations in patients with systemic sclerosis
- The effect of bosentan appears more marked in the subgroup of severe patients with more than 3 active digital ulcers at outset of treatment
- Prevention of digital ulcers with bosentan is associated with improved hand function (eating and dressing)
- Bosentan does not appear to speed healing of digital ulcers, which persisted in 50% of all subjects for up to 24 weeks
- The data from this study are congruent with an earlier randomized controlled trial
- In the absence of a clinical effect on Raynaud's phenomenon, these data support the contention that chronic endothelin receptor antagonism has an important effect on vascular integrity and function in systemic sclerosis

REFERENCES

1. Vancheeswaran R, et al. J Rheumatol 1994; 21:1268-76.
2. Korn JH, et al. Arthritis Rheum 2004; 50:3985-93.3.

ACKNOWLEDGEMENT

The study was supported by a grant from Actelion Pharmaceuticals, Allschwil, Switzerland